P1629

Gender Differences in Attitudes and Practices Towards Body Skin Care

Keith Ertel, PhD, Heather Focht, BChE, Christina Dooley, BCHE, Jamie Moak, BS
P&G Beauty & Grooming, Cincinnati, Ohio USA

INTRODUCTION

Females are the traditional focus for skin care product development and promotion, and considerable effort is placed on understanding their views on body skin condition and their body skin care needs. Male-oriented products for body skin care are a more recent focus area. Consequently, the understanding of male views on body skin care and needs is not as well developed as it is for their female counterparts.

OBJECTIVE

The objective of this work was to examine how males and females view their body skin care needs and to assess their attitudes towards treatment modalities for the identified needs.

METHODS

Research was conducted by an independent agency among a panel representative of the United States adult population comprising 303 males and 313 females. Subjects provided responses to a series of questions related to attitudes towards body skin condition, body skin care habits and practices, and attitudes towards various cosmetic interventions. Percentages were compared using a chi-square test. A 10% significance level was used for comparisons, which is standard for this type of research,

[1]2005 AAD Annual Meeting, Scientific Poster P1008

[2]Cutis 2005;76 (suppl 6):3-6

[3]2006 AAD Annual Meeting, Scientific Poster P1118

RESULTS

- Males and females agree on the top 5 qualities of healthy skin, although higher percentages of females identified these factors as being important. Skin moisturization is linked to the majority of these qualities.
- Bother from dry skin ranks high for both males and females. (Figure 1) Physical (skin type, age) and environmental (weather, wind, heating systems) factors were identified as top causes of dry skin by both sexes.
- Applying a leave-on lotion was the top dry skin intervention identified by both sexes. However, males are less likely to use hand or body leave-on lotion products than females. (Figure 2.)
- Lack of time (convenience) was the top reason given by males for not using a leave-on lotion to treat dry skin and was significantly ($P \leq 0.10$) more important to males than females. Males especially may benefit from using a product such as an in-shower body lotion that couples moisturization to an existing habit.[1]
- Additionally, expense and lack of knowledge about what product to use are frequently identified as use limiting factors by both sexes. Educating patients about cleansing and moisturizing is recognized as an important unmet patient need.[2]
- Skin-feel parameters are also more important to males than females ($P \leq 0.10$); scent is also a factor. Males may benefit from using a moisturizing body wash for increased convenience and a more tailored cleansing/moisturization experience.[3]
- Although the study showed that they are generally less body skin focused, males are more likely to seek help from a dermatologist for their dry skin than females. (Figure 2)

Top 5 Rated Qualities of Healthy Skin

Response rates between males and females all differ at $P \leq 0.10$.

	Male	Female
Soft and smooth looking	60%	80%*
Soft and smooth to the touch	59%	81%*
Moisturized, not dry and flaky	58%	82%*
Clear, not blemished	48%	71%*
Free from itch	48%	58%*

Figure 1: Bother from dry skin rated on a 0 (not at all bothered) to 5 (extremely bothered) scale.

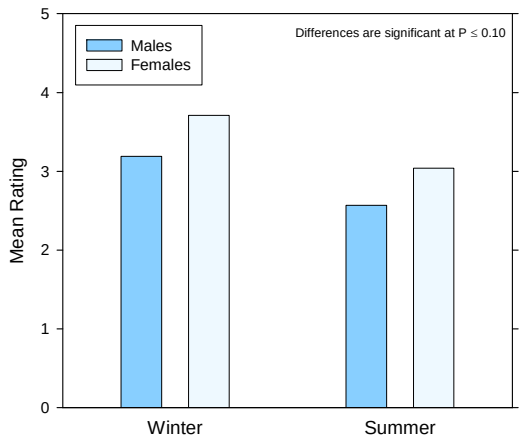
Males
Females
Differences are significant at P ≤ 0.10
Mean Rating
0
1
2
3
4
5
Winter
Summer

Figure 2: Interventions most commonly used to treat dry skin.

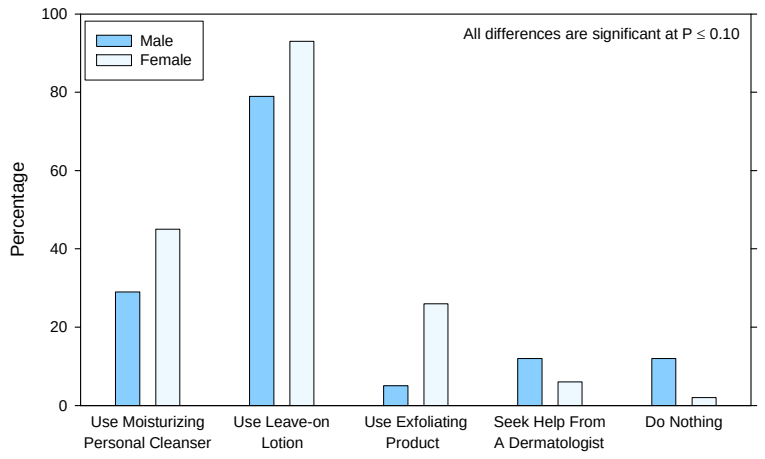
Male
Female
All differences are significant at P ≤ 0.10
Percentage
0
20
40
60
80
100
Use Moisturizing Personal Cleanser
Use Leave-on Lotion
Use Exfoliating Product
Seek Help From A Dermatologist
Do Nothing

CONCLUSIONS

Males and females share similar views on body skin care needs but males are less likely to use products to treat these needs. Dermatologists can play a role in educating their patients, especially their male patients, on the importance of body skin care and product selection. Dry skin is a common issue for both sexes, but males especially may benefit from in-shower body lotions and moisturizing body cleansers that provide convenience with good in-use aesthetics.

This work was funded by P&G Beauty & Grooming.