P1624

Potentiated Pyrithione Zinc Shampoo Usage Leads to Pronounced Alterations in Scalp Skin Histology and Biochemistry that Underlie Efficacy in Dandruff/Seborrheic Dermatitis

Kathy Kerr, B.S., Trevor Darcy, Ph.D., James Henry, Ph.D., Haruko Mizoguchi, Ph.D., James Schwartz, Ph.D., Kevin J. Mills, Ph.D.
P&G Beauty & Grooming, Cincinnati, Ohio USA

INTRODUCTION

Dandruff and scalp seborrheic dermatitis (D/SD) are inflammatory, hyper-proliferative conditions triggered at least in part by a skin response to the presence of commensal *Malassezia* yeasts. The outward manifestations of the conditions flakes, pruritus and erythema reflect histopathological and biochemical changes within scalp skin that give rise to these symptoms. Fungal control with PTZ shampoo is a valid therapeutic strategy and is known to resolve flaking symptoms, and as demonstrated by the following data an additional benefit is the normalization of the underlying skin physiology to restore a healthy scalp status.

OBJECTIVE

To histologically and biochemically characterize the status of scalp skin in D/SD, and to determine how various pathophysiological parameters change upon treatment with either a potentiated pyrithione zinc (PTZ) shampoo or placebo treatment.

METHODS

A population of severe D/SD patients was identified and treated for three weeks with either a commercial potentiated PTZ shampoo or a placebo control, which was a non-medicated commercially available shampoo. Biopsies were taken from lesional sites at baseline and the end of study for histomorphometric analysis. Scalp stratum corneum samples were also obtained for evaluation of biochemical markers of inflammation (e.g., IL-1a, IL-RA), hyper-proliferation (total protein), differentiation (Keratin 1,10,11), and barrier integrity (stratum corneum lipids, Human Serum Albumin and Involucrin protein markers). These data were then compared to reduction in flaking symptoms assessed by an expert grader to determine the effect of the treatment on these four measures of scalp health.

RESULTS

Reduction of Flaking Symptoms

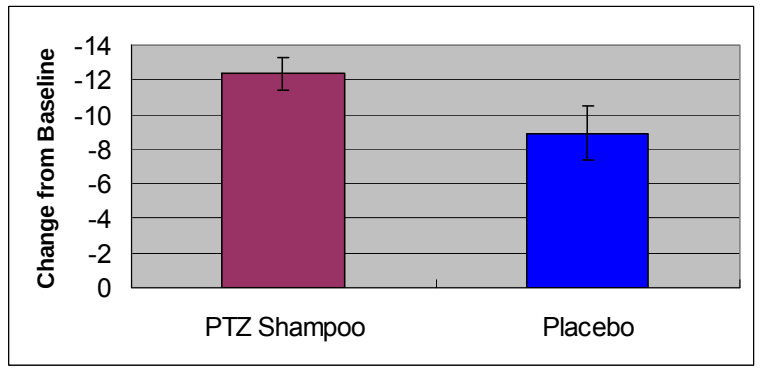
Change from Baseline
-14
-12
-10
-8
-6
-4
-2
0
PTZ Shampoo
Placebo

Treatments were statistically different, 1-sided p-value = 0.056

Histopathology

Qualitative Assessments*

Placebo

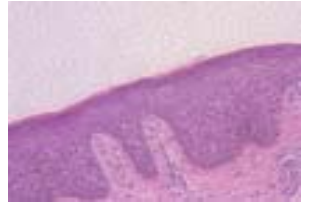

Baseline

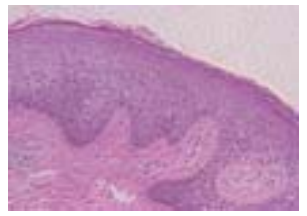

3 Weeks

PTZ Shampoo

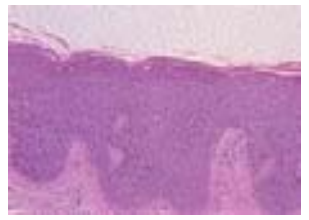

Baseline

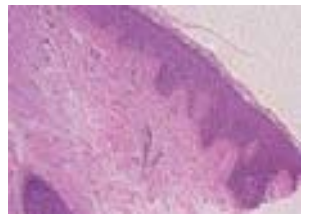

3 Weeks

* *Representative photomicrographs, all 20x magnification*

Quantitative Assessments

Treatment	Mean Epidermal Thickness, µm					Mean # Nucleated Cell Layers				
	Baseline	3 Week	*P value**	% Change	*n*	Baseline	3 Week	*P value**	% Change	*n*
Placebo Control	117.5	109.2	0.592	-7.1	15	11.2	10.5	0.623	-6.2	15
PTZ-Shampoo	125.7	102.7	0.007	-18.3	19	12.9	10.4	0.009	-19.3	19

*P Values derived from Student's *t* Test; significant difference is $p<0.05$.

Biomarkers

Proliferation (flaking)

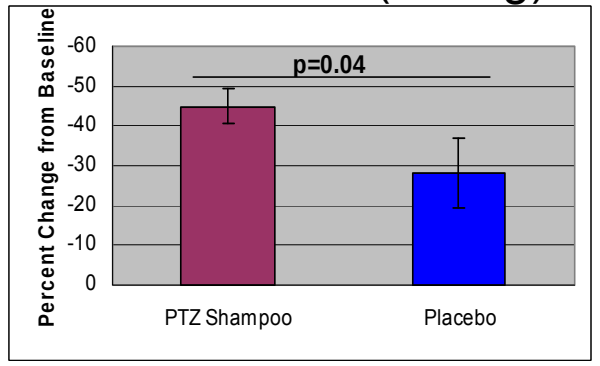
Percent Change from Baseline
-60
-50
-40
-30
-20
-10
0
p=0.04
PTZ Shampoo
Placebo

Inflammation

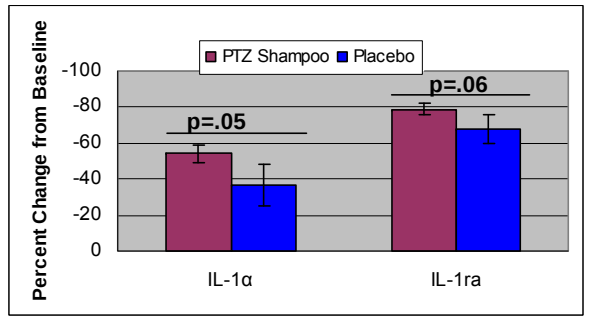
Percent Change from Baseline
PTZ Shampoo
Placebo
-100
-80
-60
-40
-20
0
p=.05
p=.06
IL-1α
IL-1ra

Differentiation

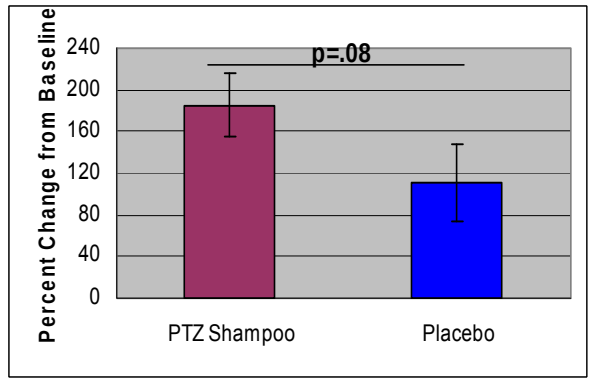
Percent Change from Baseline
240
200
160
120
80
40
0
p=.08
PTZ Shampoo
Placebo

Barrier Integrity/Repair

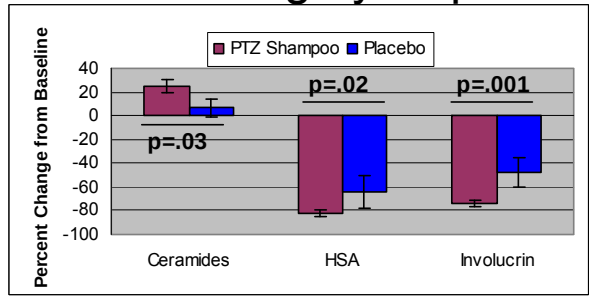
Percent Change from Baseline
PTZ Shampoo
Placebo
40
20
0
-20
-40
-60
-80
-100
p=.03
p=.02
p=.001
Ceramides
HSA
Involucrin

1-sided p-values shown for PTZ shampoo versus placebo

This work was funded by P&G Beauty & Grooming.

CONCLUSIONS

Three weeks usage of a potentiated PTZ shampoo led to a dramatic reduction in D/SD symptoms that was accompanied by significant improvements in all measured indicators of scalp health.

After treatment with the potentiated PTZ shampoo, but not the placebo, flaking and inflammatory indicators decreased significantly, cell proliferation decreased and differentiation increased indicating a restoration of equilibrium. Epidermal barrier integrity was also improved.

The resolution of hyperplasia and inflammation accompanied symptom resolution, supporting the mechanistic link between them. Effective therapies for D/SD must demonstrate treatment of both symptoms and their underlying causes to provide confidence in their value to the patient.

Reference

Warner RR, Schwartz JR, Boissy Y, et al: Dandruff has an altered stratum corneum ultrastructure that is improved with zinc pyrithione shampoo. J Am Acad Dermatol 45:897–903, 2001

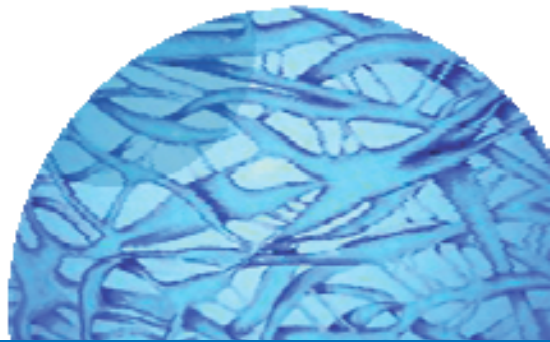