Palmitoyl-Pentapeptide: Retinol-like Efficacy with Less Irritation

Karl Lintner, Ph.D.[1]; Larry R. Robinson, Ph.D.[2]; Donald L. Bissett, Ph.D.[2]

[1]Sederma, Le Perray-en-Yvelines cedex, France; [2]The Procter & Gamble Company, Cincinnati, OH, U.S.A.

P&G BEAUTY SCIENCE

INTRODUCTION

Clinical testing of the peptide palmitoyl pentapeptide (palmitoyl-lysine-threonine-threonine-lysine-serine; pal-KTTKS) has demonstrated reduction in the appearance of wrinkles without the irritation or negative effect on skin barrier associated with a technology such as retinol (1,2). Other data (1,3) have revealed that this peptide stimulates production of dermal matrix components (in particular collagen) which may be involved in its skin activity. Based on *in vitro* and acute *in vivo* testing of pal-KTTKS, we report here additional activities which may contribute to its skin benefit effect, specifically thickening of epidermis and reduction in aging skin's excess production of dermal glycosaminoglycan (GAG).

OBJECTIVE

To determine the effect of pal-KTTKS vs. retinoid on skin effects that may be associated with improvement in aging skin: epidermal thickness and dermal GAG production.

METHODS

GAG assay

Content of hyaluronic acid (HA), as a measure of GAG, in fibroblast cell culture supernatant media was determined with a commercial HA antibody kit (Corgenic Ltd., 75 Broadway, Peterborough, PE1 1SY, United Kingdom). Fibroblasts were neonatal or from a 57 year old donor. Cells were harvested for GAG analysis when they were near confluency.

Epidermal thickness assay

Sites on the inner forearm of 12 female subjects were treated with occlusive patches containing vehicle, 0.02% (200 ppm) all-trans-retinol in vehicle, or 100 ppm pal-KTTKS in vehicle. Patches were applied to the test sites on days 1, 3, and 5. The skin was blind graded for redness (0-6 grading scale) after each patch removal and just before biopsy on day 6 for blind histological assessment (H&E). Epidermal thickness was measured by image analysis. The number of granular cell layers was counted.

RESULTS

Glycosaminoglycan (GAG) data

Elevation in GAG is a characteristic of photodamaged/wrinkled skin (4). While a low level of GAG is required for normal structure and function of the dermal matrix, excess levels are associated with poor visible appearance of skin, e.g., the wrinkled skin of Shar Pei dogs is the result of excess dermal GAG (5). In clinical testing with chemical peel (6) and in mice with topical trans-retinoic acid (7), reduction in wrinkles is also associated with reduction in dermal GAG content.

In culture, fibroblasts from an older donor produced 2-3 fold more GAG than cells from a young donor (Table I).

The peptide pal-KTTKS at extremely low doses was effective in reducing the over production of GAG by fibroblasts from an older donor, and was as effective as a much higher dose of retinoid (Figure 1). This reduction effect of retinoid has been reported previously (8).

Epidermal thickness data

In histological sections, there was significant thickening of epidermis in skin treated with either retinol or pal-KTTKS (Table II). Retinol also increased granular layer thickness, an expected retinoid effect, while peptide did not, indicating a difference in mechanism.

Redness data

Retinol was significantly irritating vs. vehicle under patch, while pal-KTTKS was not irritating (Figure 2).

Table I: Ratio of GAG production by dermal fibroblasts from old vs. young donors

Days in culture	1	2	3	4	7	11
GAG ratio (old/young)	2.30	2.64	2.68	2.83	3.05	1.98

Figure 1: Reduction in dermal GAG production by pal-KTTKS

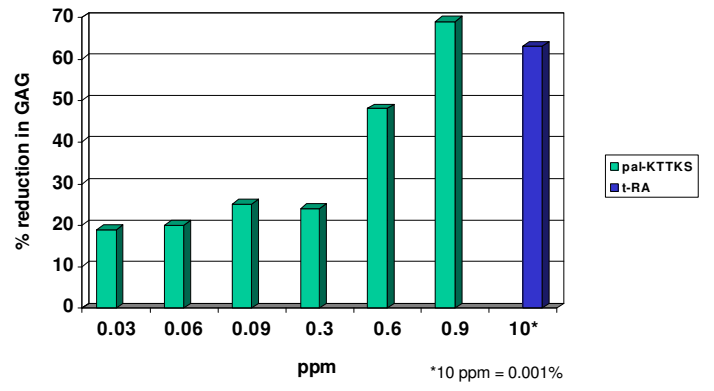
% reduction in GAG
70
60
50
40
30
20
10
0
0.03
0.06
0.09
0.3
0.6
0.9
10*
ppm
pal-KTTKS
t-RA
*10 ppm = 0.001%

Table II: Histological and image analysis observations of biopsy samples

Treatment	Epidermal thickness (microns)	Granular layer thickness (1-3 scale)
Vehicle	52.98	1.59
0.02% (200 ppm) retinol	62.60 ($p < 0.05$)	2.36 ($p < 0.05$)
100 ppm pal-KTTKS	60.97 ($p < 0.05$)	1.73

Figure 2: Irritation scores under patch

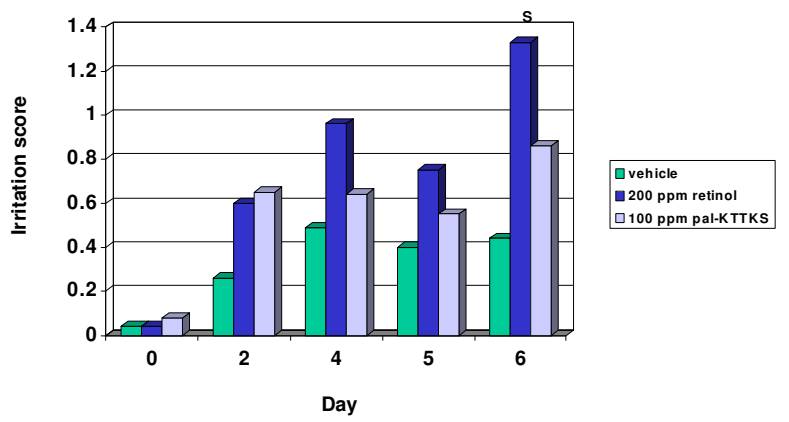
Irritation score
1.4
1.2
1
0.8
0.6
0.4
0.2
0
s
0
2
4
5
6
Day
vehicle
200 ppm retinol
100 ppm pal-KTTKS

CONCLUSIONS

1. **The peptide pal-KTTKS shares some activities with retinoids which may contribute to its observed activity in reducing the appearance of facial wrinkles:**
 - **reduces excess dermal GAG found in older skin**
 - **increases epidermal thickness**
2. **In contrast to retinol, pal-KTTKS did not induce irritation in forearm patch testing even at much higher levels than the 3 ppm dose shown previously to improve the appearance of facial skin wrinkles (1,2). Additional facial appearance improvement data are presented at this meeting (9).**

References

1. C. Mas-Chamberlin et al., Poster #160, World Congress of Dermatology, 2002.
2. L. R. Robinson et al., Poster #179, World Congress of Dermatology, 2002.
3. C. Mas-Chamberlin et al., Poster #438, World Congress of Dermatology, 2002.
4. S. Conzalez, et al., Photochem. Photobiol., 70, 248-253 (1999).
5. R. W. Dunstan et al., Handbook of Mouse Mutations with Skin and Hair Abnormalities, pp. 524-525 (1994).
6. A. M. Kligman et al., Plast. Recon. Surg., 75, 652-659 (1975).
7. E. Schwartz et al., J. Invest. Derm., 104, 518-522 (1995).
8. T. J. Smith, J. Clin. Endocrin. Met., 70, 655-660 (1990).
9. D. L. Bissett et al., Poster #307, AAD Annual Meeting, 2003.

THIS STUDY WAS FUNDED BY THE PROCTER & GAMBLE CO.